Discovery and optimization of potent and selective inhibitors of USP7 to enhance anti-tumor immunity and target tumor growth

Betty Abraham, Lavanya Adusumilli, Berenger Biannic, Delia Bradford, Dirk Brockstedt, Martin Brovarney, David Chian, Christophe Colas, Gene Cutler, Xinping Han, Dennis Hu, Scott Jacobson, Sherra Johnson, Aparna Jorapur, Paul Kassner, Deepika Kaveri, John Ketcham, Andrea Kim, Paul Leger, Lisa Marshall, Sachie Marubayashi, Jack Maung, Jenny McKinnell, Cesar Meleza, Andrew Napper, Yamini Ohol, Akinori Okano, Leanne Peiser, Deepa Pookot, Payal Rana, Jerick Sanchez, Jacob Schwarz, Nick Shah, Grant Shibuya, Michael Sun, Silpa Suthram, Oezcan Talay, Angela Wadsworth, David Wustrow, Kyle Young; ***FLX Bio, Inc. South San Francisco, CA***

Abstract

USP7 is a deubiquitinase (DUB) that regulates the levels of multiple proteins with roles in cancer progression and immune response. USP7 has been reported to stabilize oncogenes such as MDM2, destabilize and inactivate the tumor suppressors p53 and PTEN, impart resistance to DNA-damaging chemotherapy by stabilizing proteins involved in DNA damage responses, and enhance the suppressive function of regulatory T cells (T_{reg}) by stabilizing the transcription factor FOXP3. We have designed USP7 inhibitors (USP7i) that are highly potent in biochemical and cellular assays and selective for USP7 over all other DUBs. Our inhibitors bind USP7 and reduce viability of the MM.1S tumor cell line, relieve mouse T_{reg} suppression of $CD8^+$ cell proliferation in vitro, and enhance inflammation in the contact hypersensitivity model *in vivo*. In addition, our compounds are orally bioavailable with moderate permeability and low clearance.

Multiple Beneficial Mechanisms of USP7 Inhibitors

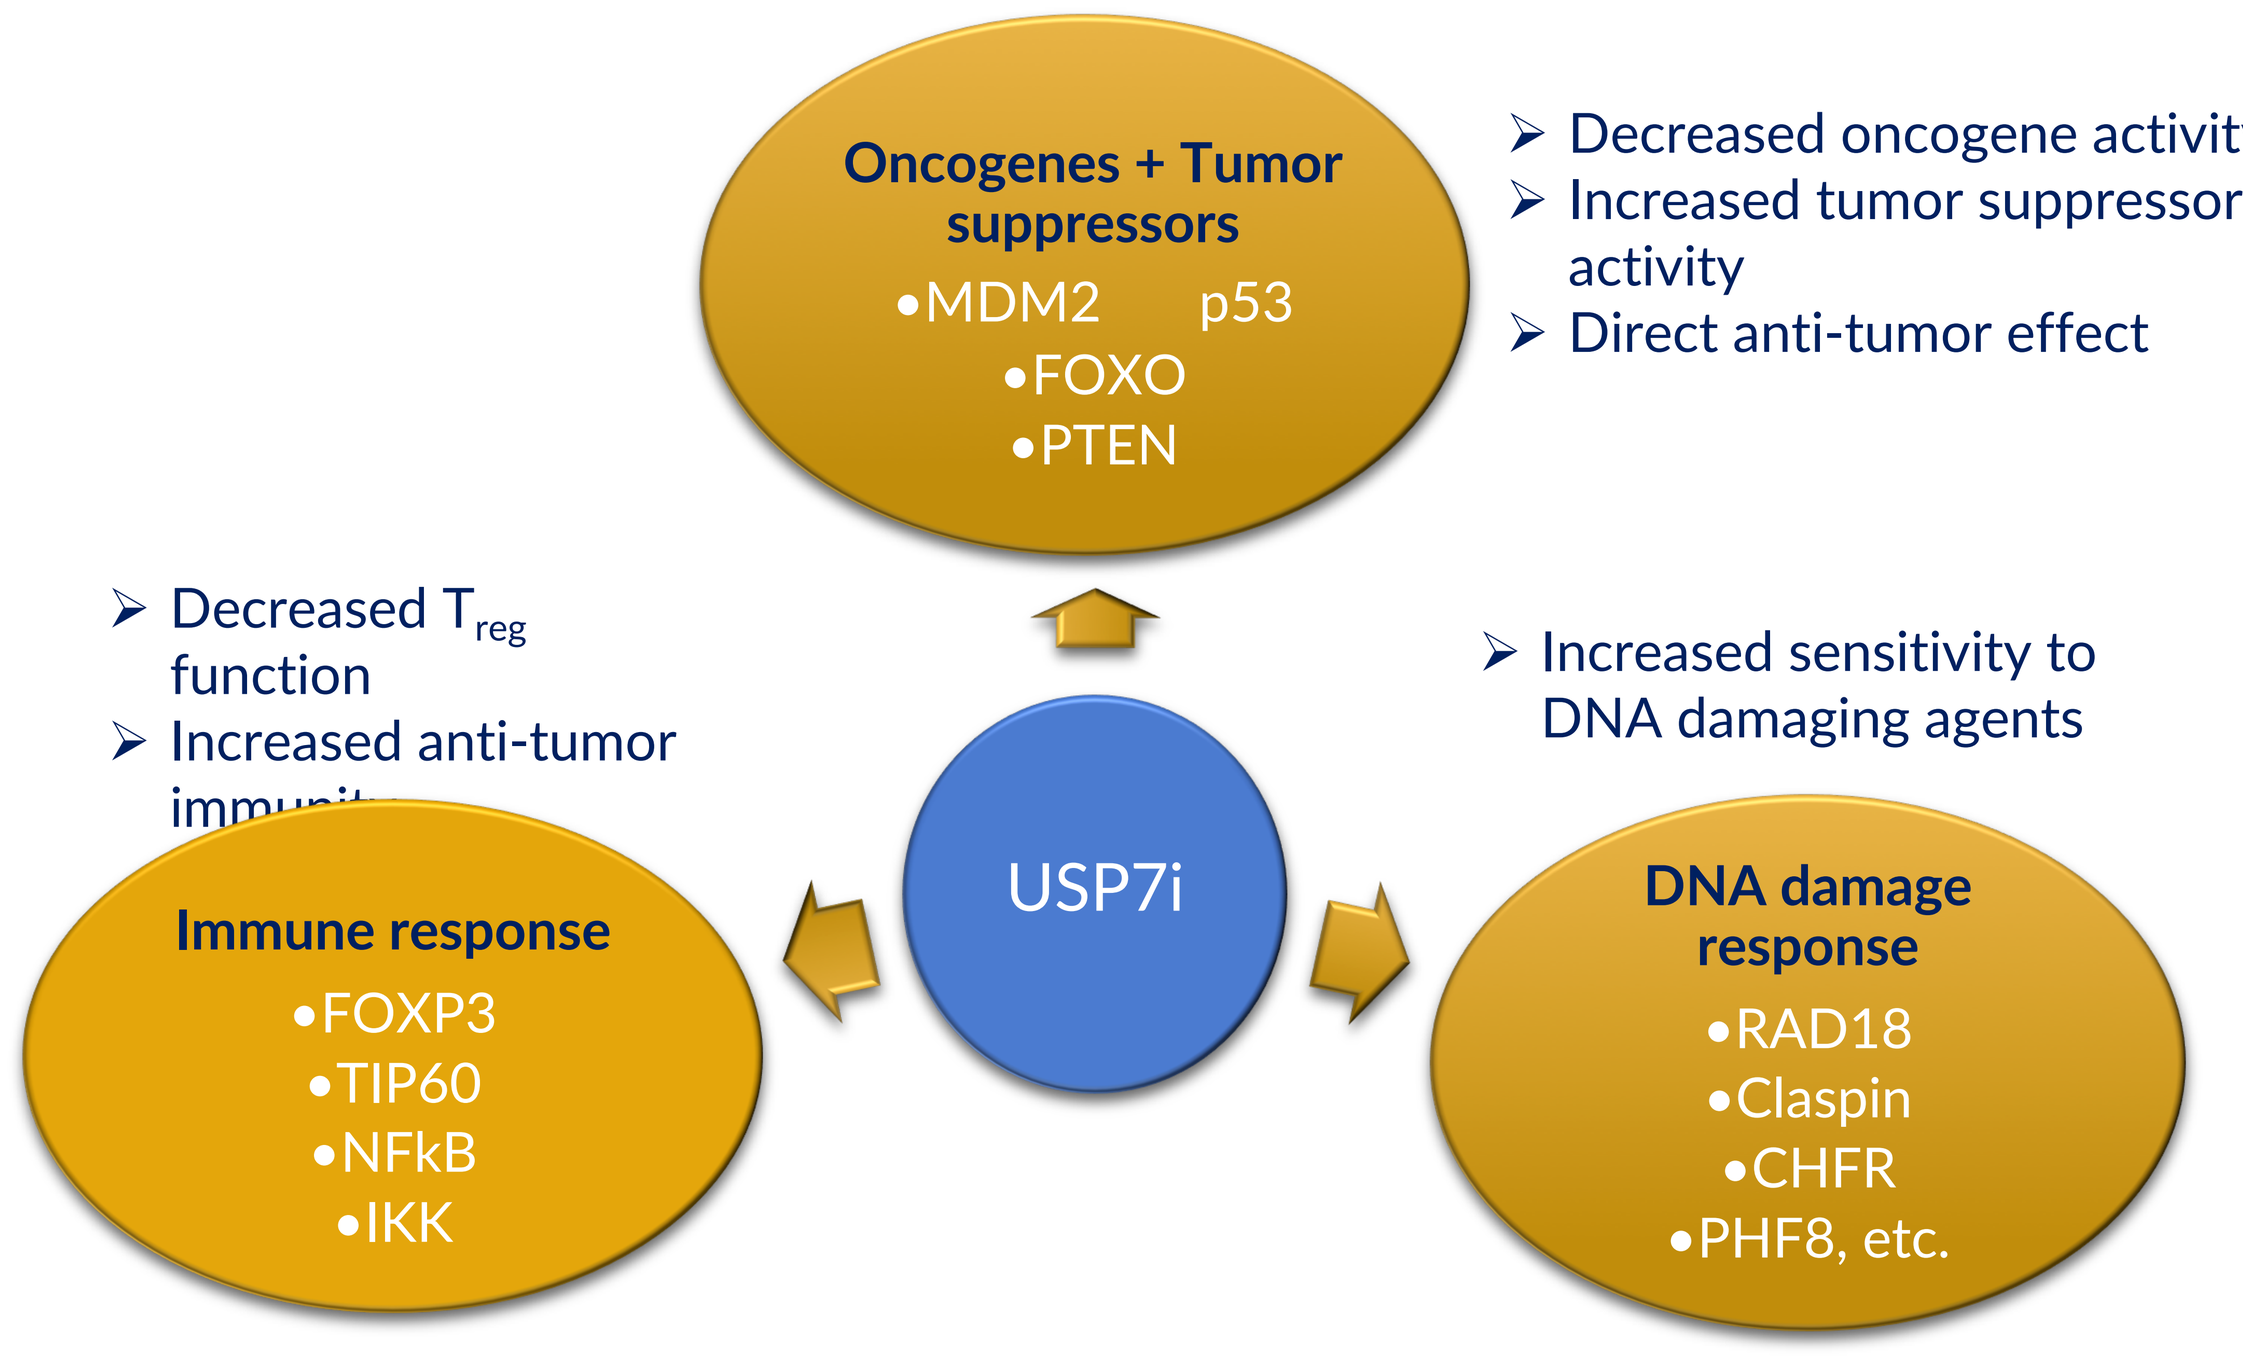
Oncogenes + Tumor suppressors
•MDM2 p53
•FOXO
•PTEN
Decreased oncogene activity
Increased tumor suppressor activity
Direct anti-tumor effect
USP7i
Decreased T_{reg} function
Increased anti-tumor immunity
Immune response
•FOXP3
•TIP60
•NFkB
•IKK
Increased sensitivity to DNA damaging agents
DNA damage response
•RAD18
•Claspin
•CHFR
•PHF8, etc.

Biochemical Potency of Compounds Used in These Studies

USP7i	USP7 IC_{50}
FLX-A	< 10 nM
FLX-B	< 10 nM
FLX-C	< 1 nM
FLX-D	< 10 nM
FLX-E	< 1 nM

1 Novel USP7 Inhibitors are Highly Selective and Potent in Biochemical and Cellular Assays

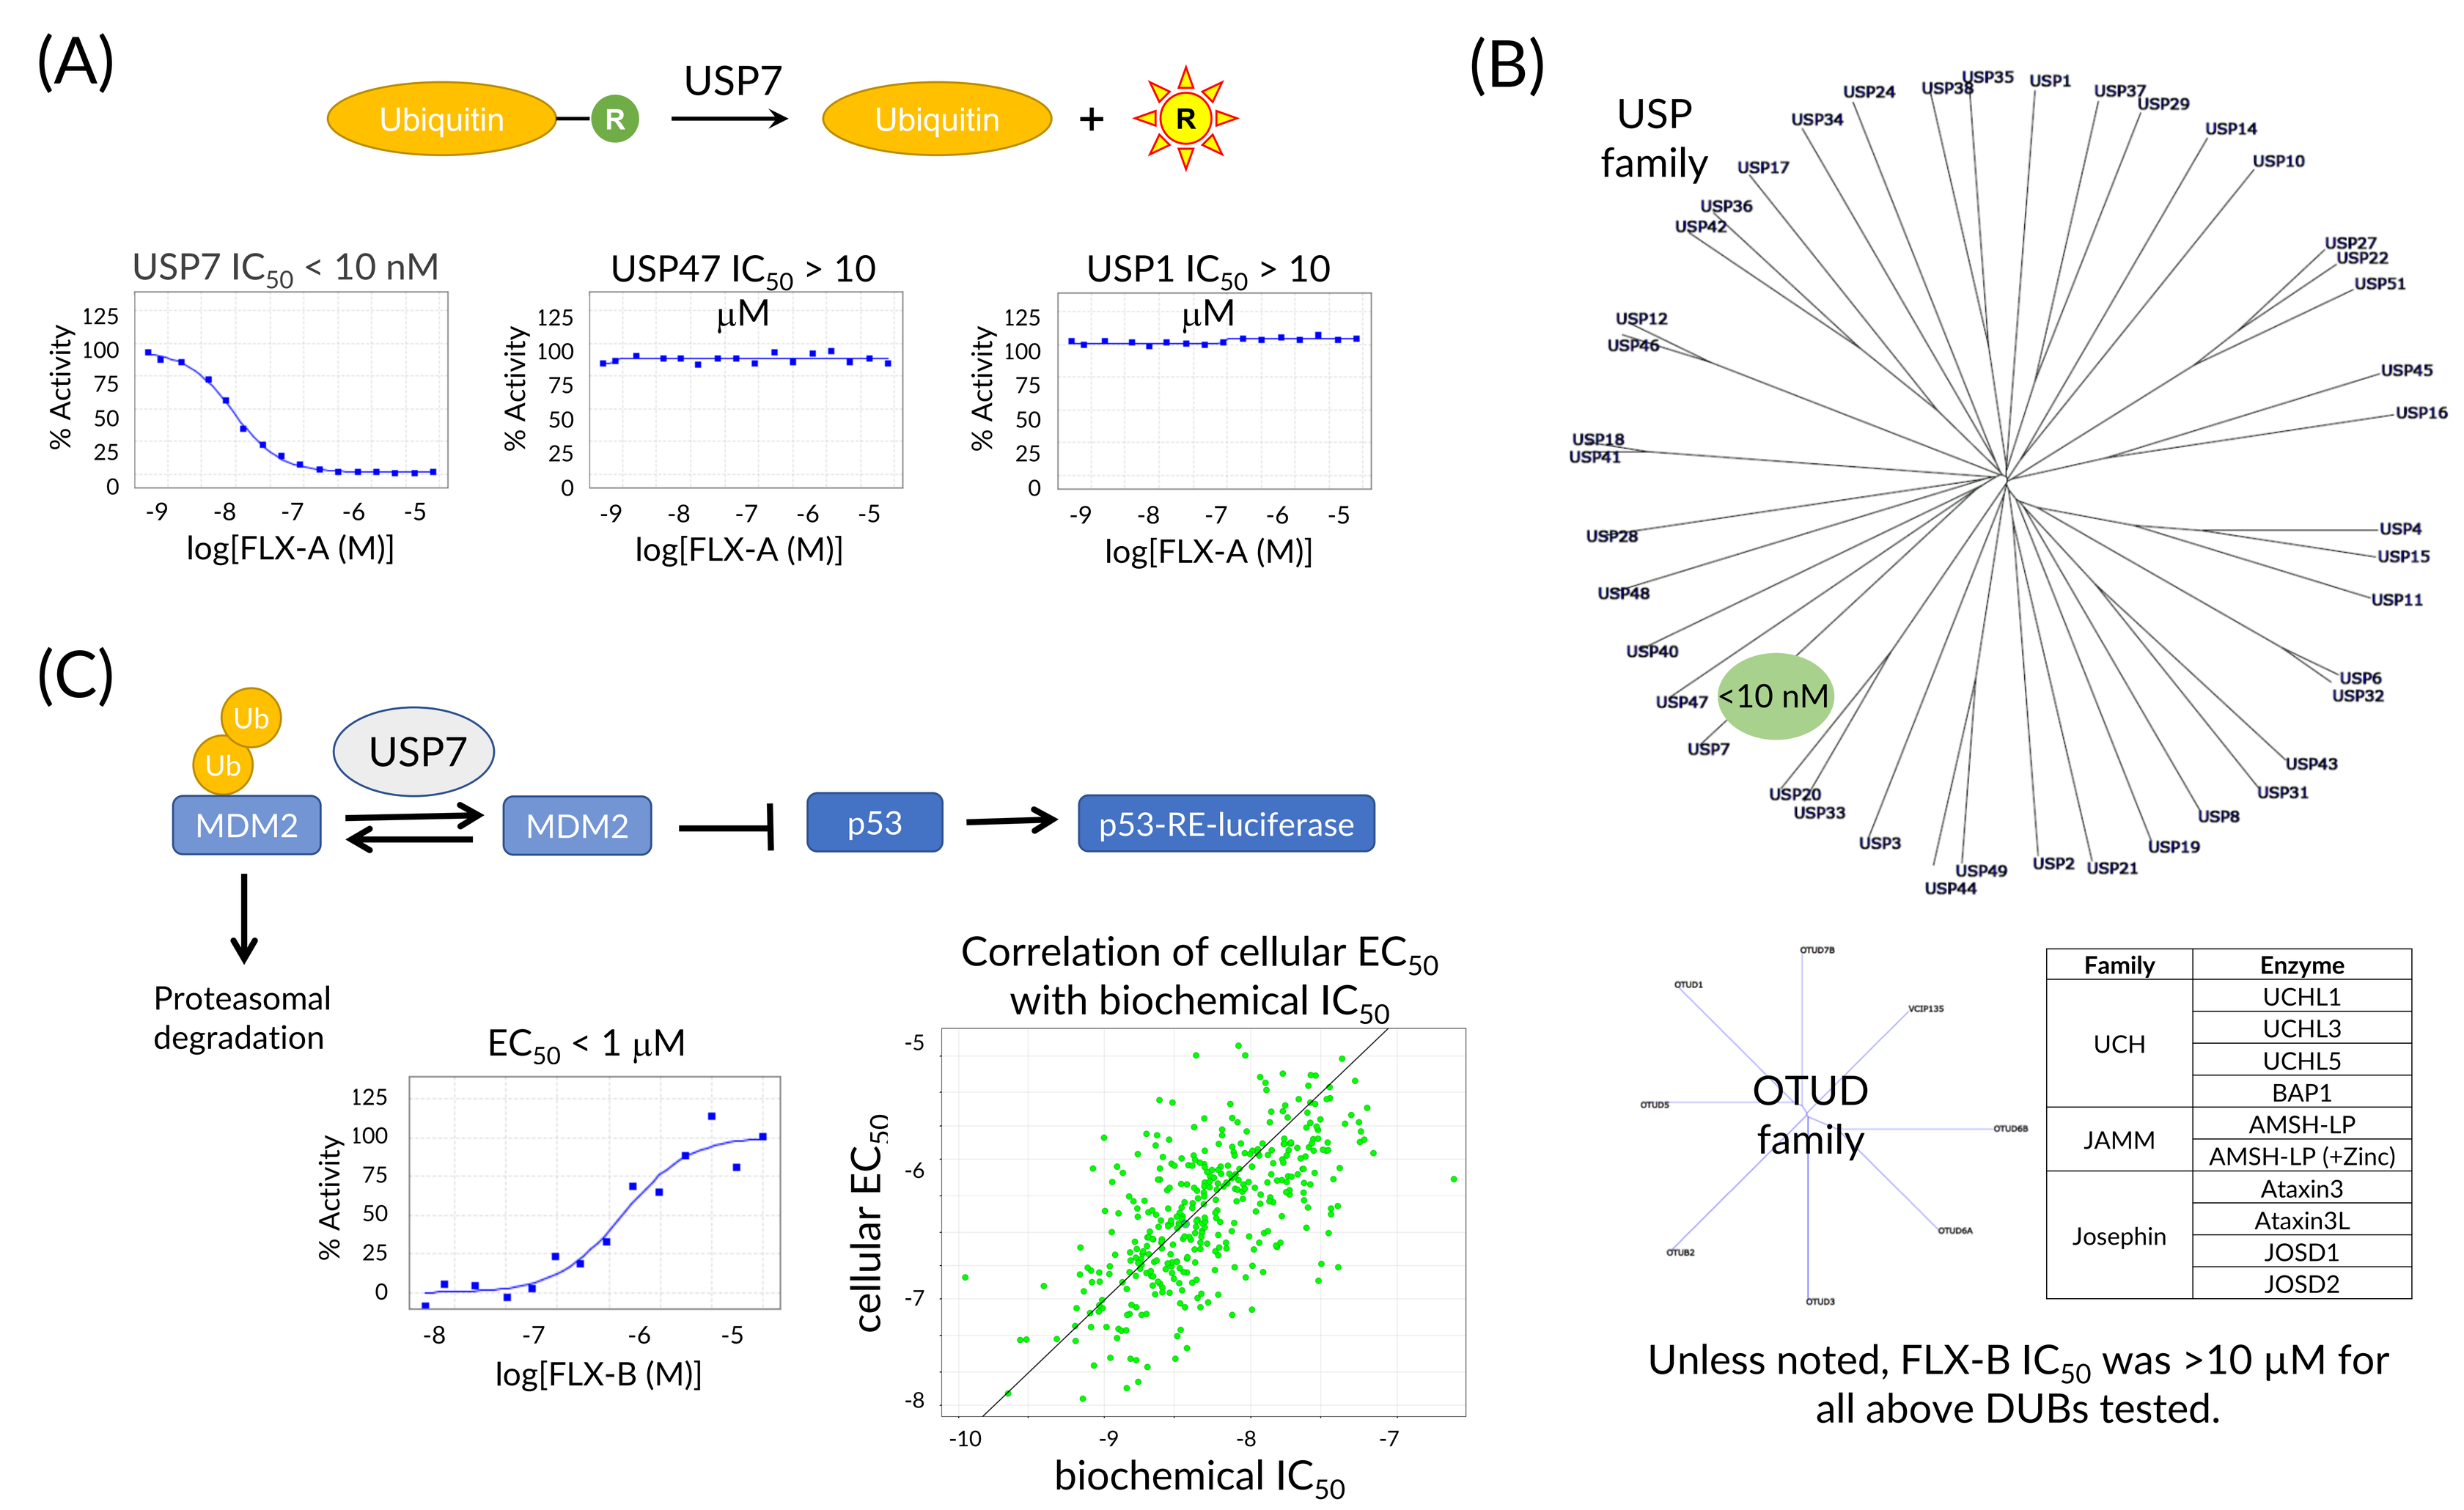
(A)
Ubiquitin
R
USP7
Ubiquitin
+
R
USP7 IC_{50} < 10 nM
USP47 IC_{50} > 10 µM
USP1 IC_{50} > 10 µM
% Activity
log[FLX-A (M)]
(B)
USP family
<10 nM
OTUD family
Unless noted, FLX-B IC_{50} was >10 µM for all above DUBs tested.
(C)
Ub
USP7
MDM2
MDM2
p53
p53-RE-luciferase
Proteasomal degradation
EC_{50} < 1 µM
log[FLX-B (M)]
Correlation of cellular EC_{50} with biochemical IC_{50}
cellular EC_{50}
biochemical IC_{50}

Family	Enzyme
UCH	UCHL1
	UCHL3
	UCHL5
	BAP1
JAMM	AMSH-LP
	AMSH-LP (+Zinc)
Josephin	Ataxin3
	Ataxin3L
	JOSD1
	JOSD2

(**A**) Biochemical activity of FLX USP7 inhibitor (USP7i) against USP7 and selectivity over USP47 and USP1 in an assay using rhodamine-labeled ubiquitin. (**B**) Selectivity of FLX USP7i over all other DUBs. (**C**) Cellular activity of FLX USP7i in a luciferase reporter gene assay of p53 activation.

2 Novel USP7 Inhibitors Show Target Engagement and Tumor Cell Line Cytotoxicity Predicted by p53 Status

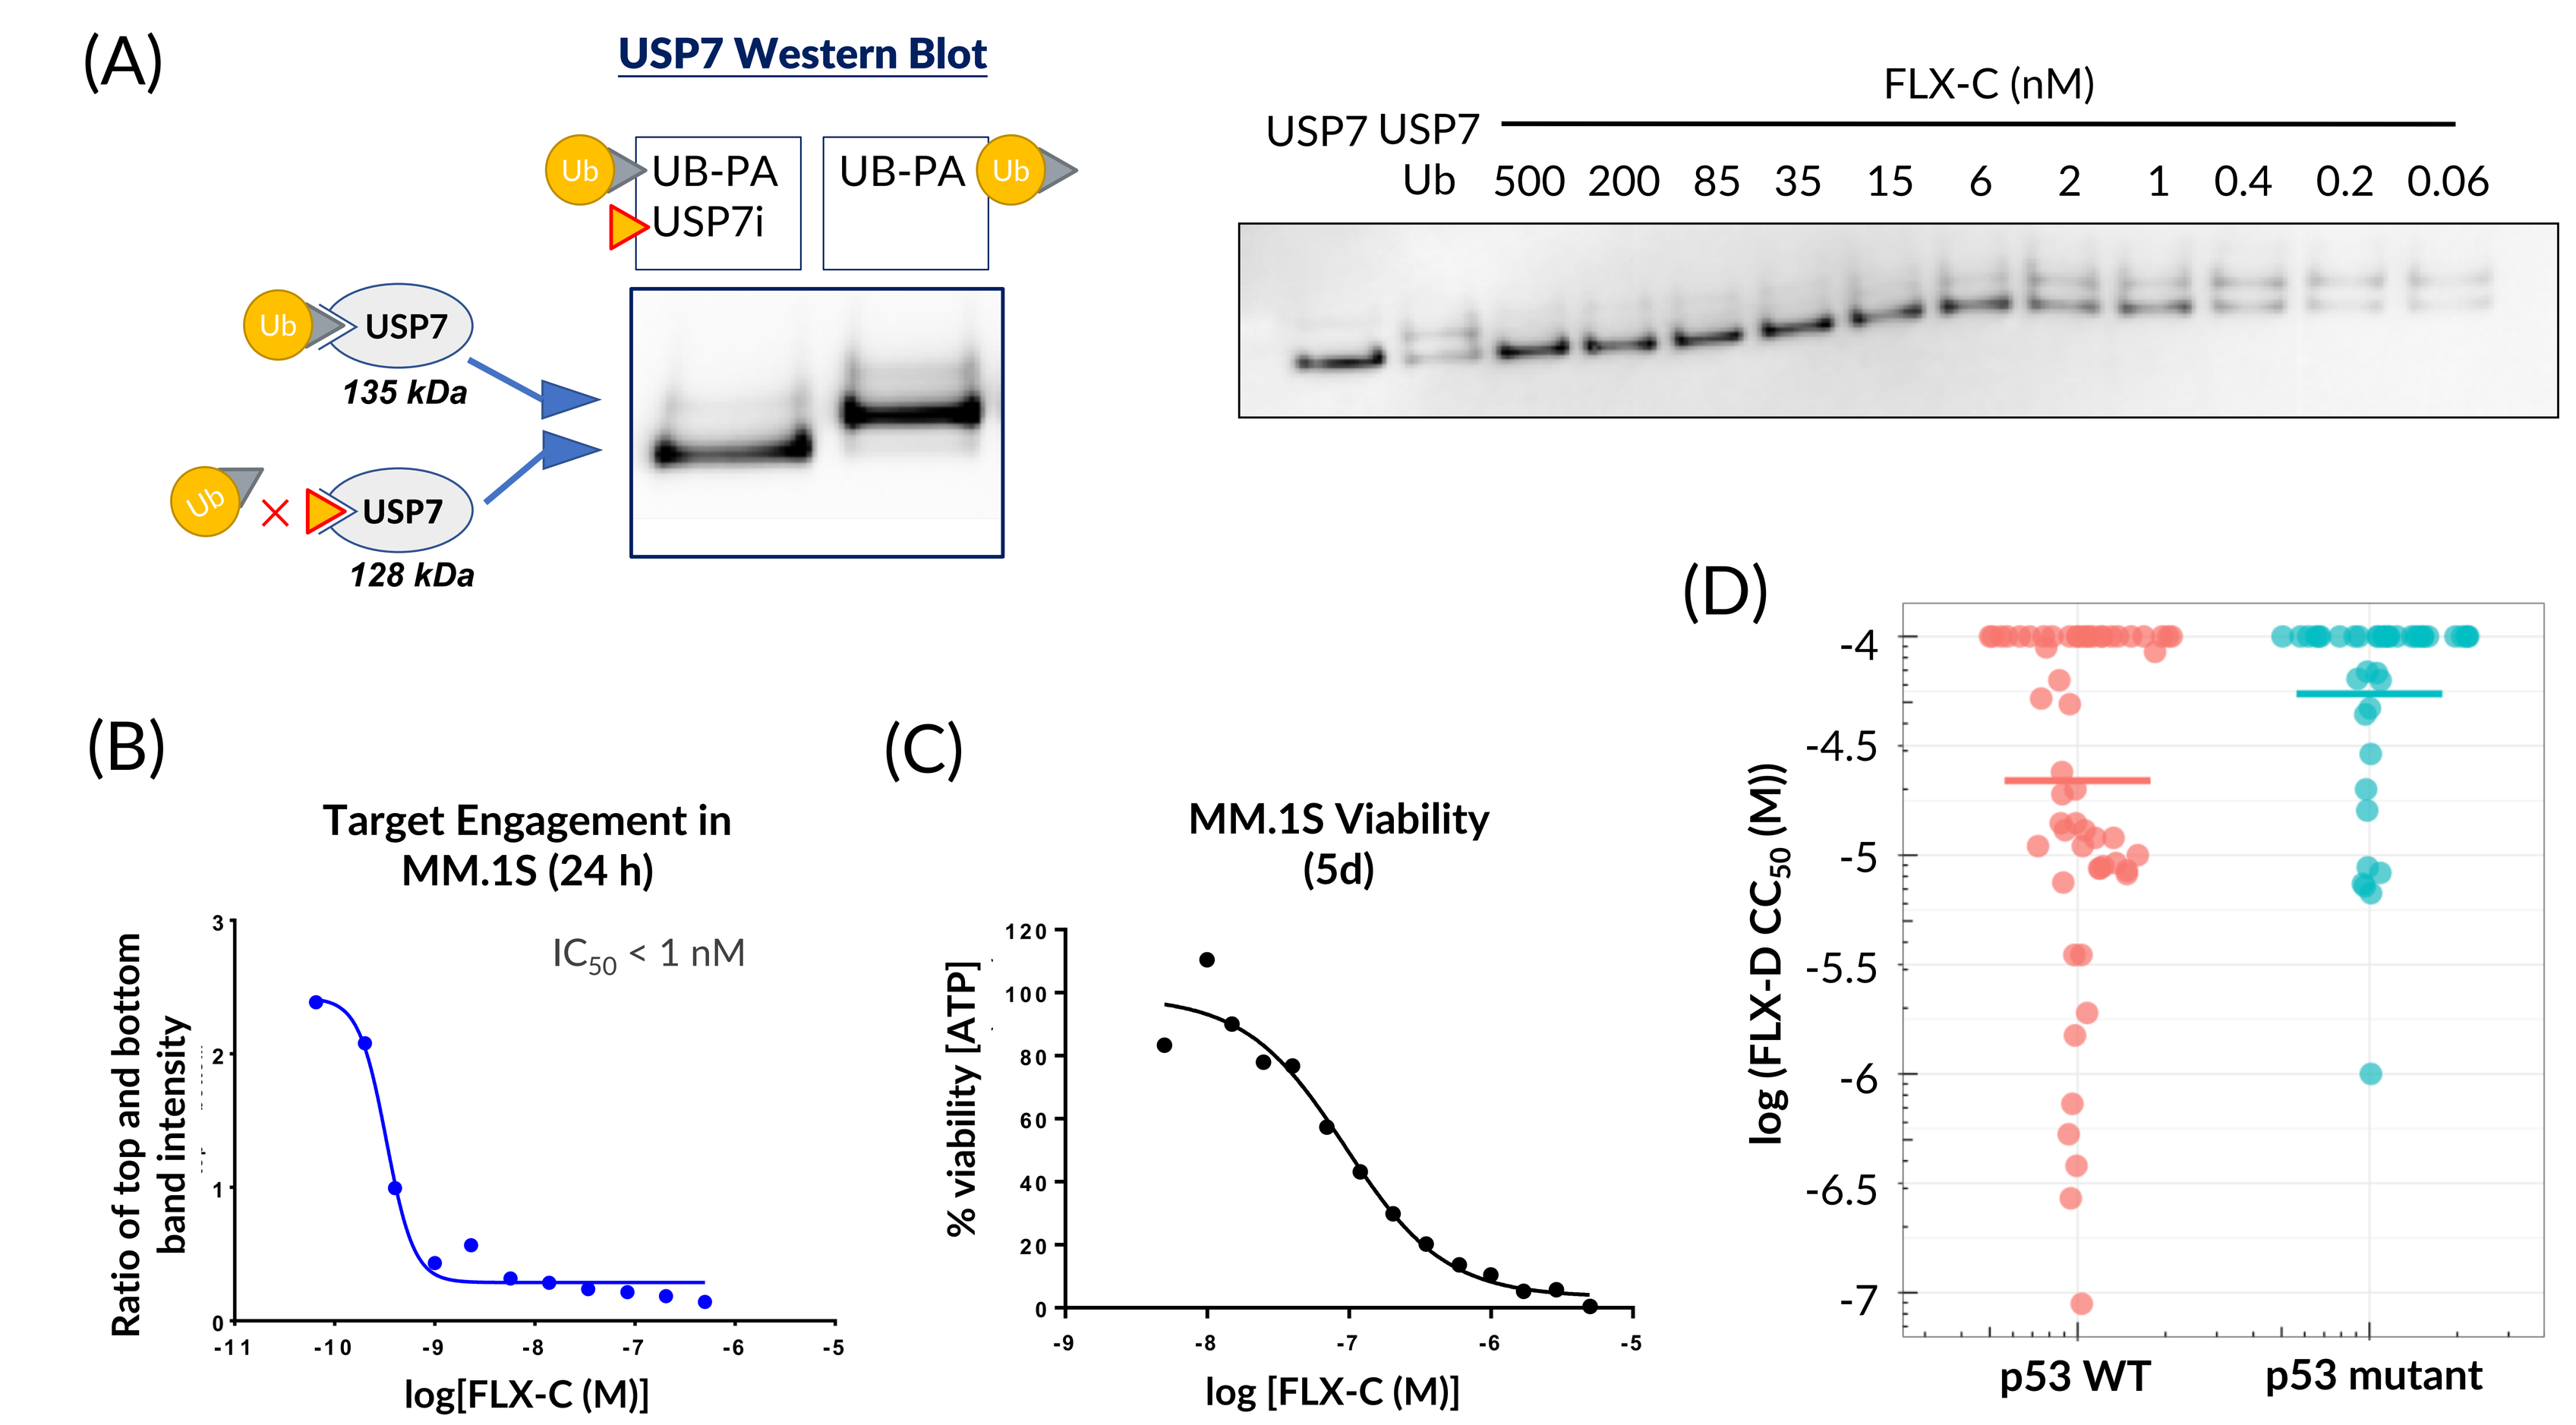
(A)
USP7 Western Blot
UB-PA
USP7i
UB-PA
USP7
135 kDa
USP7
128 kDa
FLX-C (nM)
USP7 USP7 Ub 500 200 85 35 15 6 2 1 0.4 0.2 0.06
(B)
Target Engagement in MM.1S (24 h)
IC_{50} < 1 nM
Ratio of top and bottom band intensity
log[FLX-C (M)]
(C)
MM.1S Viability (5d)
% viability [ATP]
log [FLX-C (M)]
(D)
log (FLX-D CC_{50} (M))
p53 WT
p53 mutant

(**A**) Schematic of target engagement assay and a representative Western blot. Treatment of MM.1S cells with FLX USP7i results in target engagement as measured at 24 hr (**B**) and dose-dependent cytotoxicity in a 5-day assay (**C**). (**D**) Analysis of mutational predictors of sensitivity of a panel of 100 cell lines to FLX USP7i indicated that p53 status was an important predictor of sensitivity.

3 USP7 Inhibitors Relieve Mouse T_{reg} Suppression of $CD8^+$ T Cell Proliferation

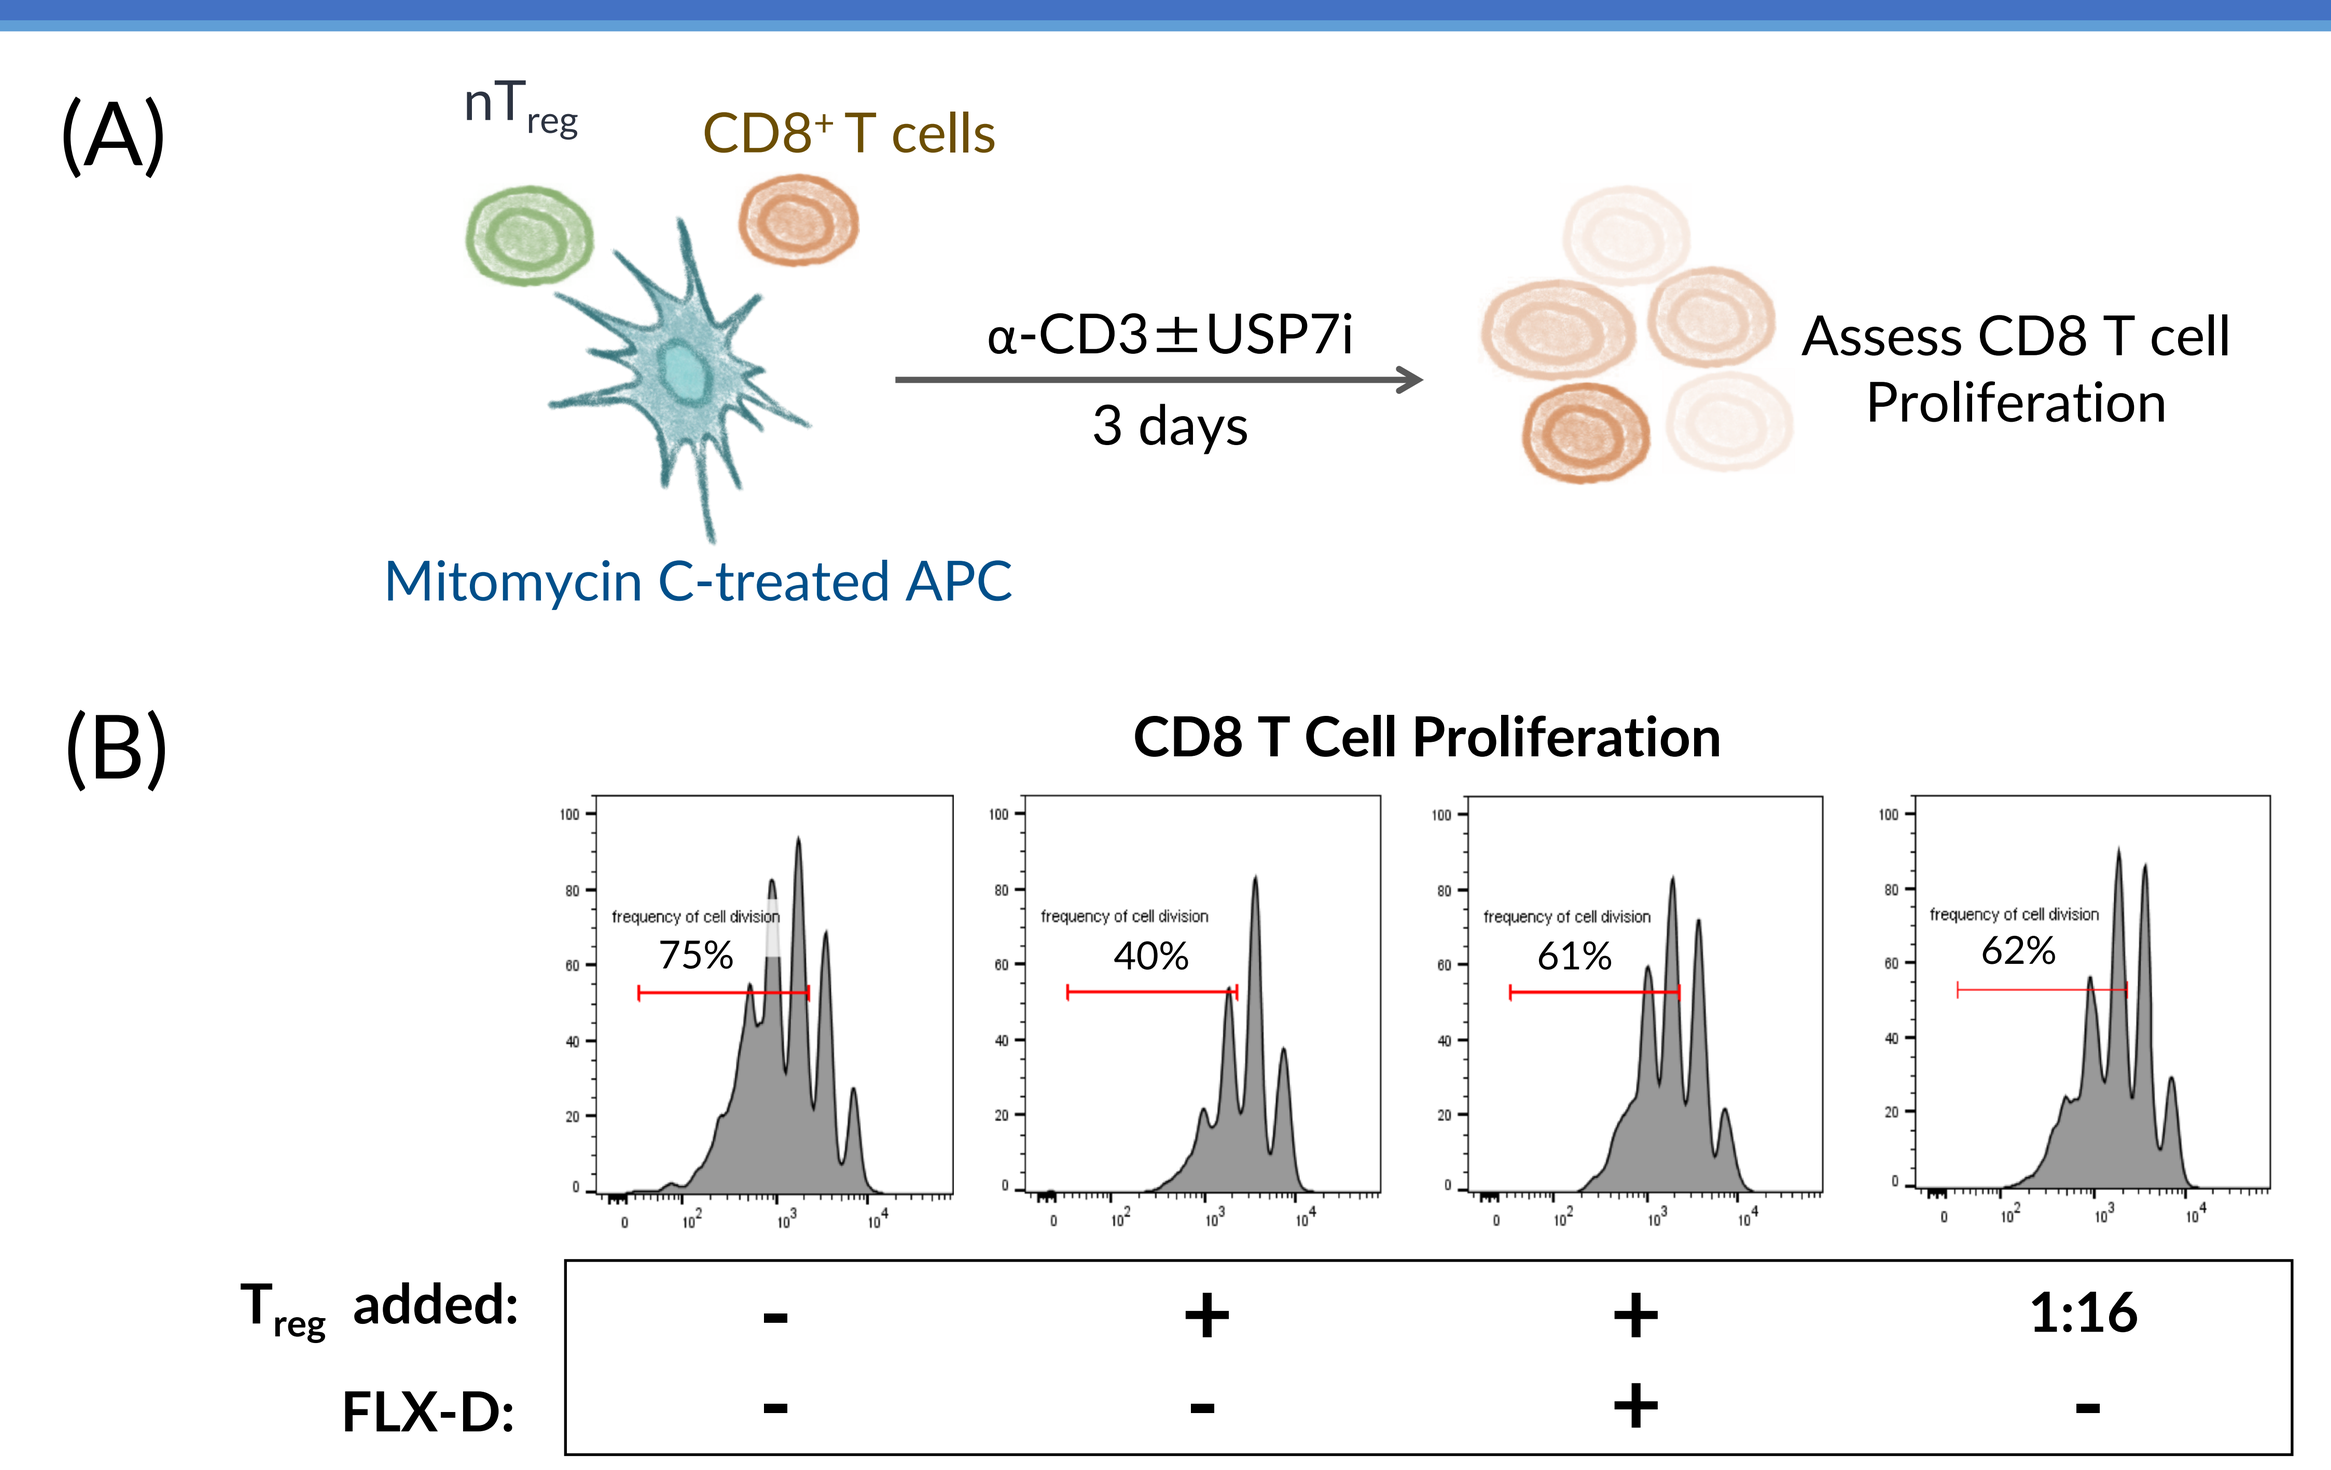
(A)
nT_{reg}
$CD8^+$ T cells
α-CD3±USP7i
3 days
Assess CD8 T cell Proliferation
Mitomycin C-treated APC
(B)
CD8 T Cell Proliferation
75%
40%
61%
62%
T_{reg} added: - + + 1:16
FLX-D: - - + -

(**A**) $CD8^+$ T cells were co-cultured with natural T_{reg} cells and antigen-presenting cells in the presence of FLX USP7i for 4 days, after which $CD8^+$ cell proliferation was assessed by flow cytometry. (**B**) FLX USP7i reverses suppression of $CD8^+$ T cell proliferation by ~40% (equivalent to diluting T_{reg} by 16-fold).

4 USP7 Inhibitors Enhance Inflammation in Contact Hypersensitivity Model *in Vivo*

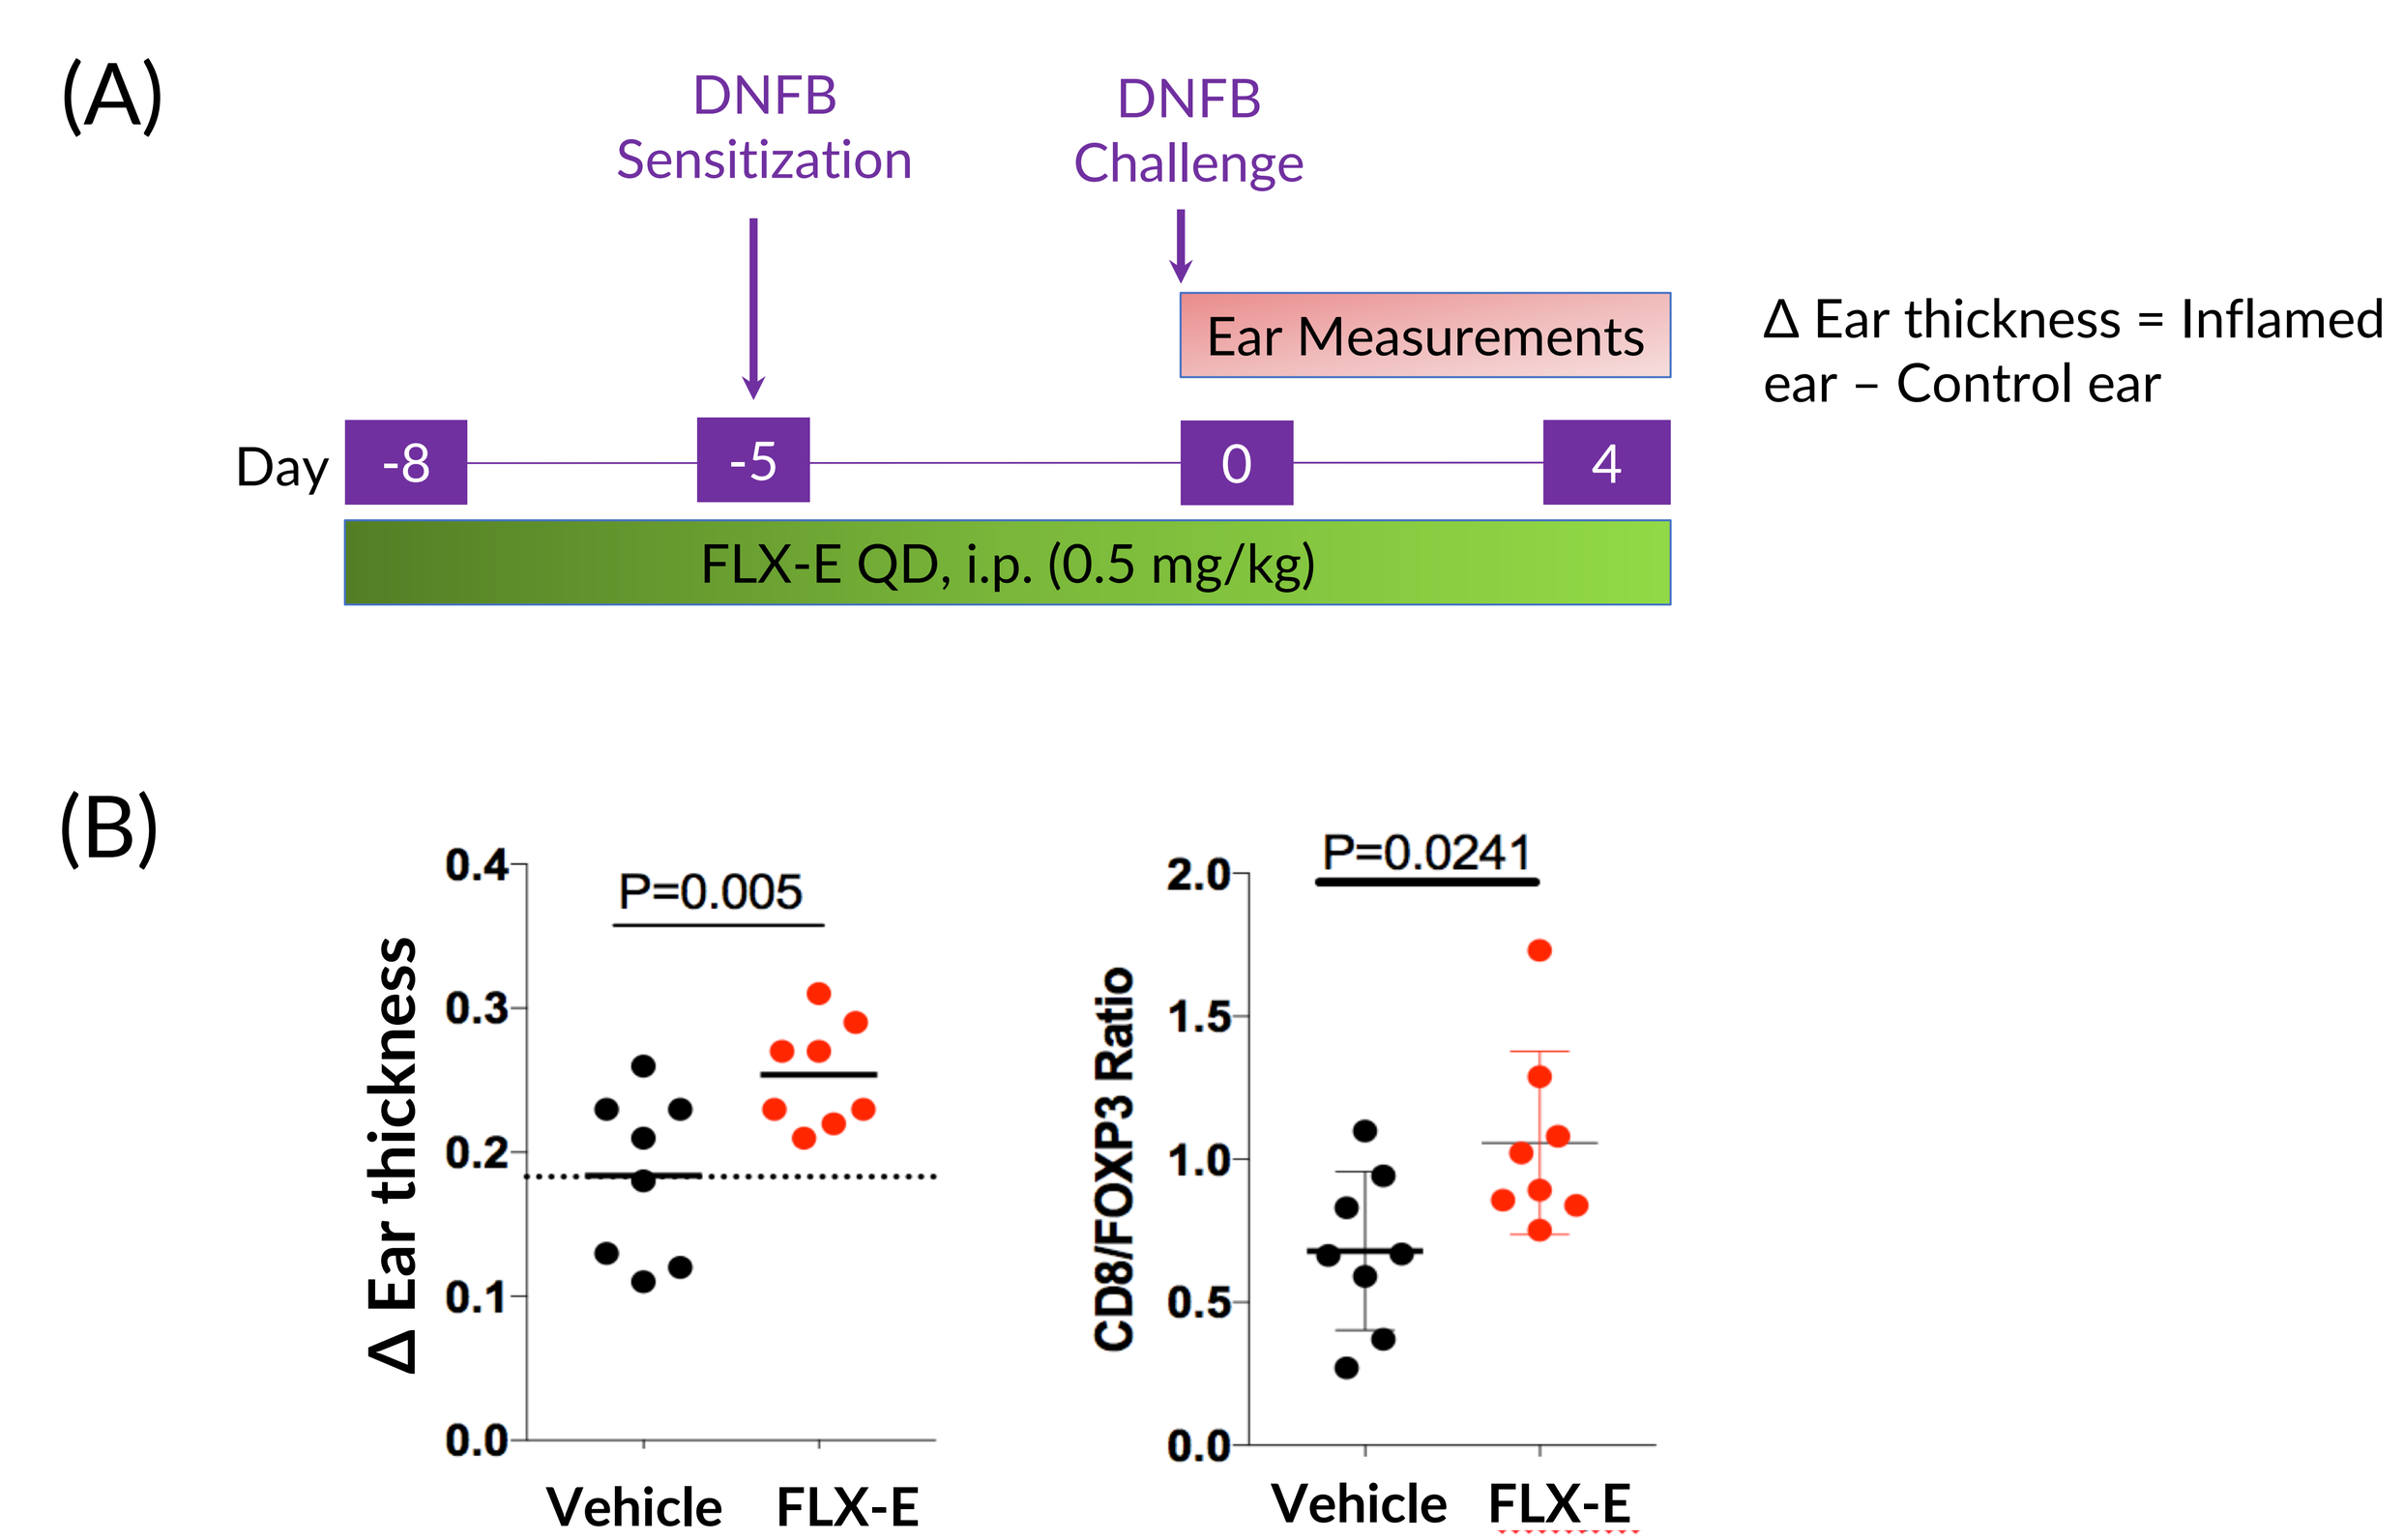
(A)
DNFB Sensitization
DNFB Challenge
Ear Measurements
Δ Ear thickness = Inflamed ear – Control ear
Day -8 -5 0 4
FLX-E QD, i.p. (0.5 mg/kg)
(B)
P=0.005
Δ Ear thickness
P=0.0241
CD8/FOXP3 Ratio
Vehicle
FLX-E

(**A**) Schematic of the contact hypersensitivity model: mice are sensitized to and later challenged with a hapten, DNFB (1-fluoro-2,4-dinitrobenzene). T_{reg} function in both sensitization and challenge phases. (**B**) FLX USP7i increases inflammation as measured by changes in ear thickness and in CD8/FOXP3 ratio.

5 USP7 Inhibitors are Highly Bioavailable

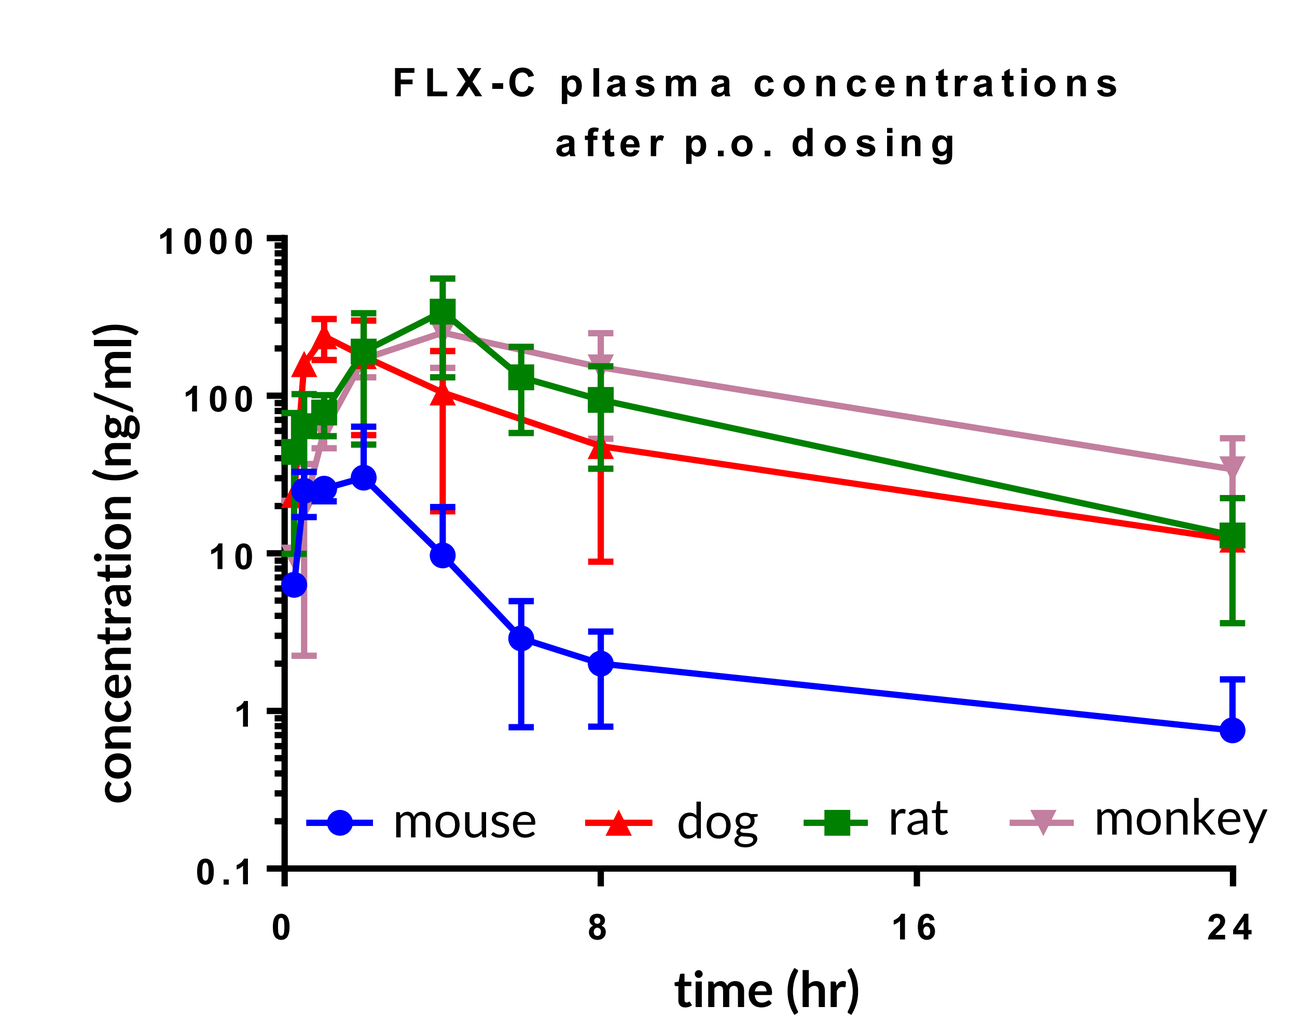
FLX-C plasma concentrations after p.o. dosing
concentration (ng/ml)
time (hr)
mouse
dog
rat
monkey

	mouse	rat	dog	monkey
% F	6	12	62	88

FLX USP7i have desirable bioavailability, allowing the possibility of oral dosing in additional pharmacological models.

Conclusions

- We have developed a novel series of potent, highly selective small molecule USP7 inhibitors with activity both *in vitro* and *in vivo*.
- FLX USP7i activate p53 in cell-based assays and are cytotoxic to the MM.1S tumor cell line *in vitro*.
- FLX USP7i increase inflammation and change CD8/FOXP3 ratio *in vivo*, indicating immune activation.
- **We believe our USP7 inhibitors are novel immunomodulators with multiple beneficial mechanisms of action including direct killing of tumor cells.**